Tumor Radiosensitization Using a Novel Metalloporphyrin in Mice Bearing SCCVII Squamous Cell Carcinomas

C Beattie[1], M Muira[2], M S Makar[2], P L Micca[2]

[1]MorEx Development Partners LLP, London, United Kingdom, [2]Brookhaven National Laboratories, Upton, NY

Introduction

MTL-005, a boronated metalloporphyrin, is being developed as a radiosensitizer. The biodistribution and efficacy of the compound has previously been studied in an EMT-6 mammary carcinoma model. Radiotherapy is commonly used to treat head and neck cancers of which over 90% are squamous cell carcinomas. The SCCVII murine squamous cell carcinoma model is a well established model for human squamous cell carcinoma, and is known to be non-immunogenic and resistant to radiation and chemotherapy.[1,2] The radiosensitizing effect and tissue distribution of MTL-005 was studied in mice implanted with SCCVII subjected to single-fraction X-ray therapy.

Materials & Methods

Animal Tumor Model

SCCVII murine squamous cell carcinoma cells were cultured in D-MEM enriched with 10% fetal bovine serum, 1% penicillin/streptomycin, and 1% L-glutamine. Cells (2 x 10^5 in 0.05 mL of medium) were then implanted sc into the left thighs of 20-25 g female C3H mice (Charles River Laboratories, Wilmington, MA).

Formulation

MTL005 was used to make three 15 mL batches of ~3 mg/mL formulations. The final concentration assayed by HPLC was 2.80 mg/mL MTL005 with 97% purity by HPLC.

Boron and MTL005 Analysis

Direct current plasma-atomic emission spectroscopy [DCP-AES] (ARL/Fisons Model SS-7) was used (detection limit: 0.1 µg B/mL) to determine boron concentrations in tissues of individual mice. Samples (50-130 mg) were digested at 60°C with sulfuric acid:nitric acid (1:1). Triton X-100 and water were added to give final concentrations of ~50 mg tissue/mL, 15% total acid v/v and 5% Triton X-100 v/v. [3] The reference standard for MTL005 was assayed using prompt-gamma spectroscopy at the Massachusetts Institute of Technology Reactor Prompt-Gamma Neutron Activation Facility.[4] The concentrations of MTL005 in the SCVII tumor and normal tissues can be calculated from the boron concentration of the porphyrin (22.5% boron). Previous work has shown that the ether linkages between the porphyrin and carborane cages remain intact in vivo.

Administration Protocol and Total doses

A total dose of 150 mg/kg MTL005 was administered to mice using three i.p. injections over an 8-hour period at 4-h intervals using 0.018 mL/gbw for each injection. Due to the large number of animals to be dosed and irradiated, it was necessary to split the animals at each X-ray dose level into two equally sized groups: Groups A and B. Animals in each Group A were injected on day 7 after tumor implantation, while animals in each Group B were injected on day 8 post tumor implantation.

Irradiations

Irradiation was carried out 1 day after the final injection of MTL005.
The animals in each Group A were irradiated 8 days, and those in Groups B were irradiated 9 days after tumor implantation. Mice were anesthetized (sodium pentobarbital, ~60 µg/g i.p.) and positioned for irradiation with the tumor-bearing leg extended across a 2-cm diameter collimated irradiation port. Tumors were irradiated using a single dose fraction of either 25 Gy or 32 Gy at a dose-rate of 2.0 Gy/min using a Philips RT-100 set, operating at 100 kVp and 8 mA.

Dosimetry

Dosimetry for x-irradiation was carried out using a thimble ionization chamber applying the 1996 IPEB code of practice. [5]

Assessments

Tumors were measured 2-3 times per week and the mice were killed humanely when the calculated tumour volume ($x^2y/2$, where x is the shorter surface dimension) exceeded 500 mm^3. All surviving mice were euthanized at day 62 or 61 post irradiation for Groups A and B respectively.
A group of five mice, each of which were given MTL005, was euthanized at the time of irradiation (24-hour clearance) for boron biodistribution data in tumor, blood, liver, spleen, brain, and skin. The liver and tumor of irradiated mice given MTL-005, were also assayed for boron concentration at the time of euthanasia.

Results

Radiotherapy

MTL-005 did not enhance overall survival in mice in the 25 Gy groups (Figure 1). Two mice in the 25 Gy only group survived to day 62 with no observable tumor (controlled tumor). The only mouse in the 25 Gy plus MTL-005 group that survived to day 62 had detectable tumor (Table 1). Mice treated with 32 Gy with or without MTL-005 had better survival than those that received 25 Gy. Four mice that received 32 Gy only survived to day 62 and 3 of these had controlled tumors. In the 32 Gy plus MTL-005 group, 8 mice survived to day 62, of these 6 had controlled tumors.

Figure 1

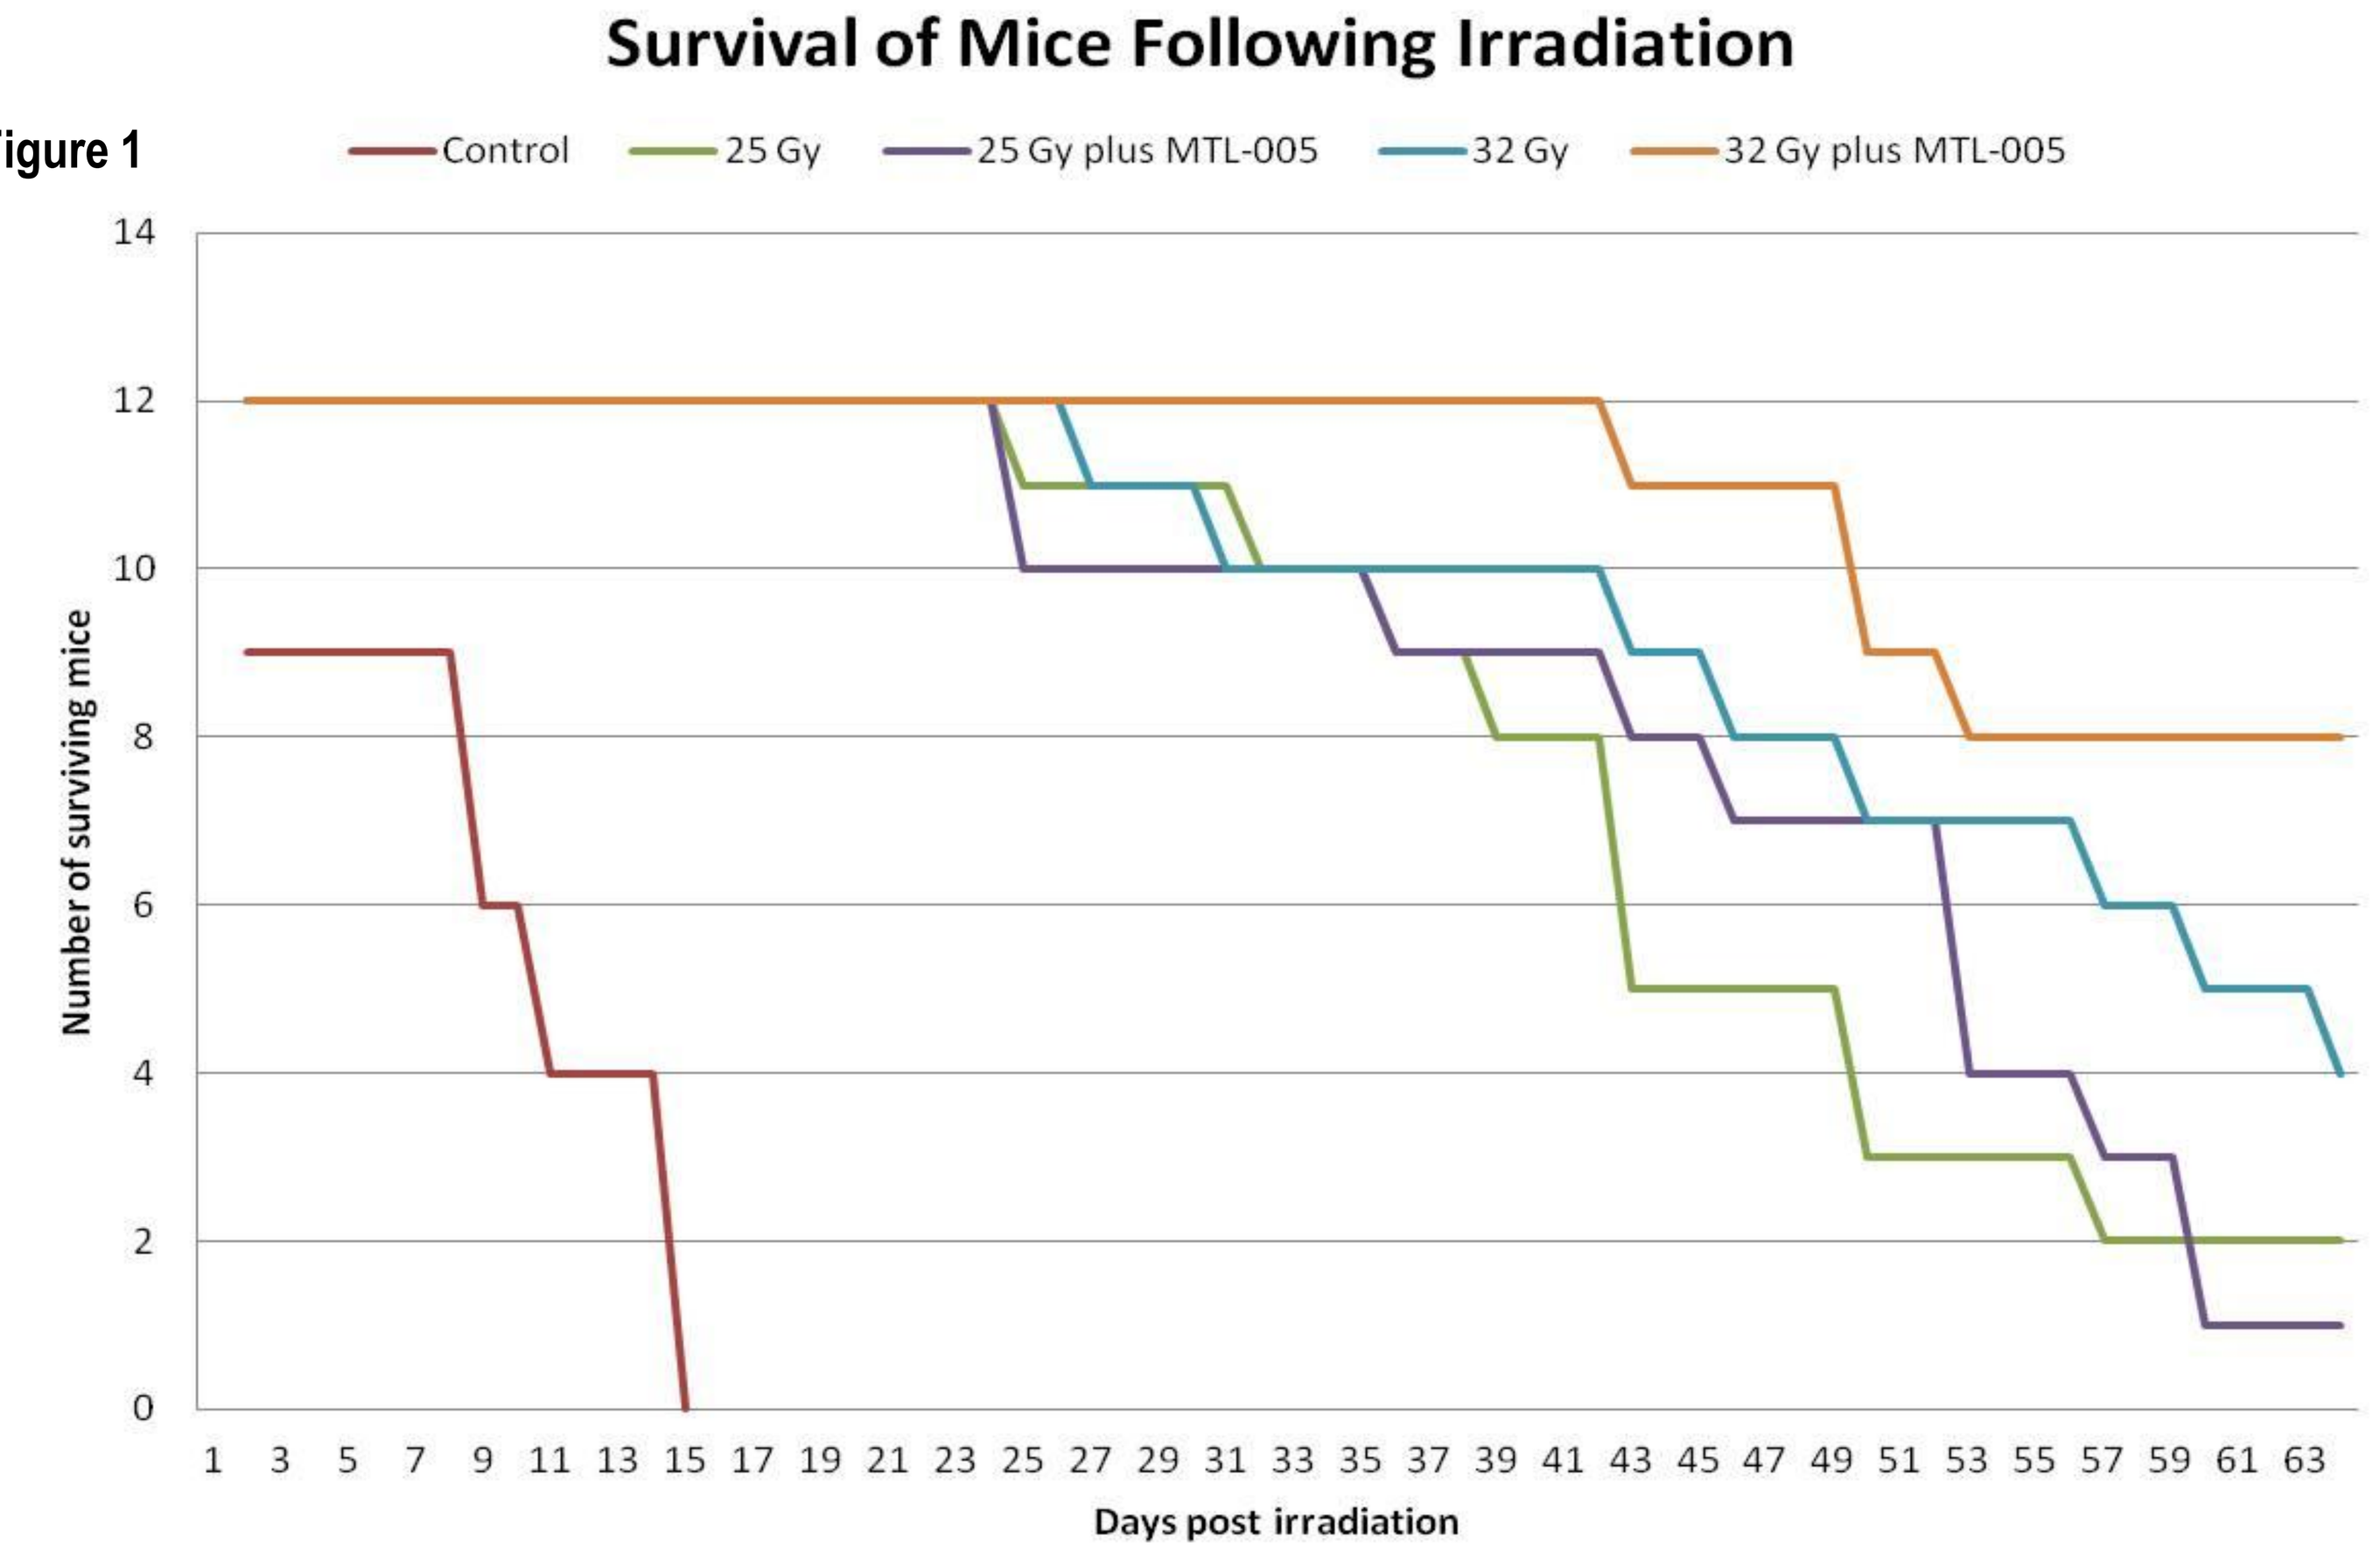
Survival of Mice Following Irradiation
Control
25 Gy
25 Gy plus MTL-005
32 Gy
32 Gy plus MTL-005
Number of surviving mice
Days post irradiation

Table 1

	Untreated Controls	25 Gy Only	25 Gy plus MTL-005	32 Gy Only	32 Gy plus MTL-005
Total Mice Irradiated	9	12	12	12	12*
Controlled Tumors at 62 days**	0	2	0	3	6
Mice alive at 62 days with observable tumors	0	0	1	1	2
%Controlled Tumors	0	16.7	0	25	54.5*

*One mouse with no observable tumour was euthanized at day 48 due to leg damage and was not counted as controlled. Controlled % based on 11 mice. **Controlled tumors defined as no observable volume.

Fig. 2

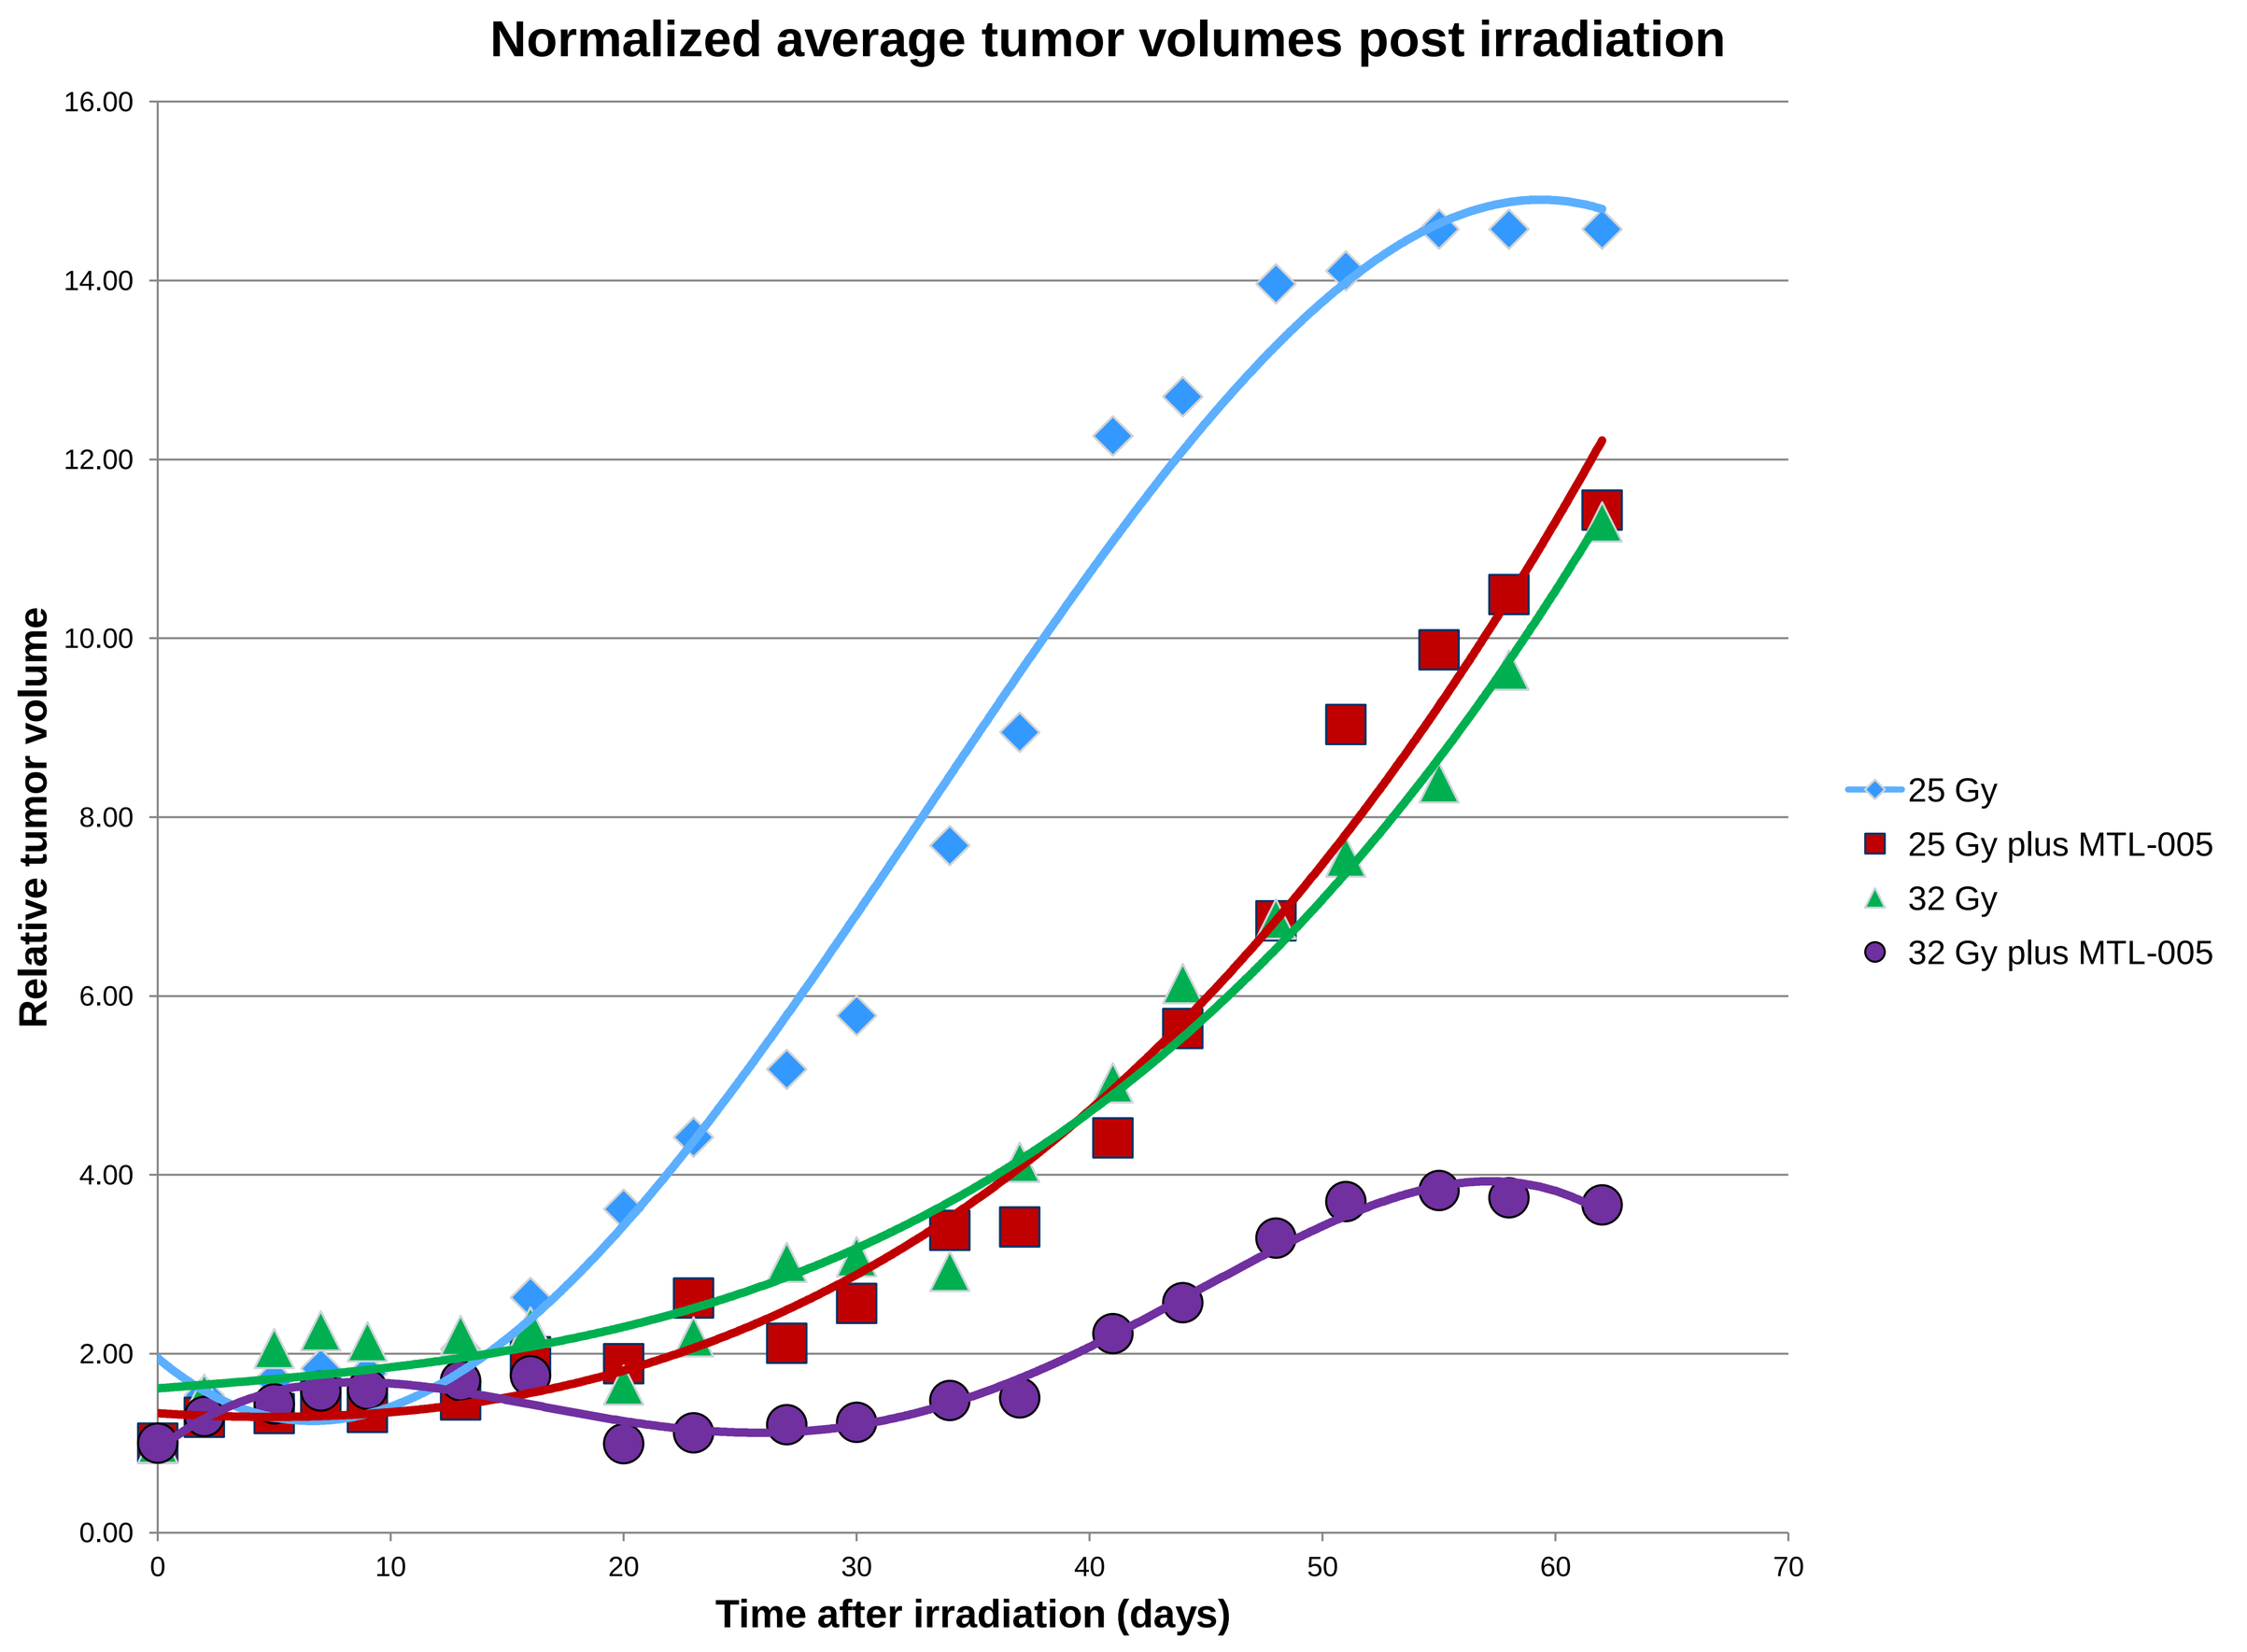
Normalized average tumor volumes post irradiation
Relative tumor volume
Time after irradiation (days)
25 Gy
25 Gy plus MTL-005
32 Gy
32 Gy plus MTL-005

Increasing the dose of radiation from 25 Gy to 32 Gy appeared to retard tumor growth (Figure 2). At each radiation dose, this effect was increased by the addition of MTL-005. The addition of MLT-005 to 25 Gy appeared to have the same effect as irradiation with 32 Gy alone. The normalized tumor doubling times in days to first animal sacrifice were 2.1; 10.5; 16.2; 18.2. and 35.1 for mice in the untreated control, 25 Gy, 25 Gy plus MTL-005, 32 Gy and 32 Gy plus MTL-005 groups respectively

Biodistribution

At 1 day post administration MTL-005 accumulated in the following tissues (figures given as µg boron/g tissue): liver (263 ± 32); spleen (100± 18); tumor (83 ± 17); blood (45 ± 12); skin (10 ± 2); brain (0.8 ± 0.4).

Boron was detected in both the liver and tumors (where present) of animals at the time of euthanasia at all time points up to and including 62 days post irradiation. (Figures 3 & 4).

Figure 3

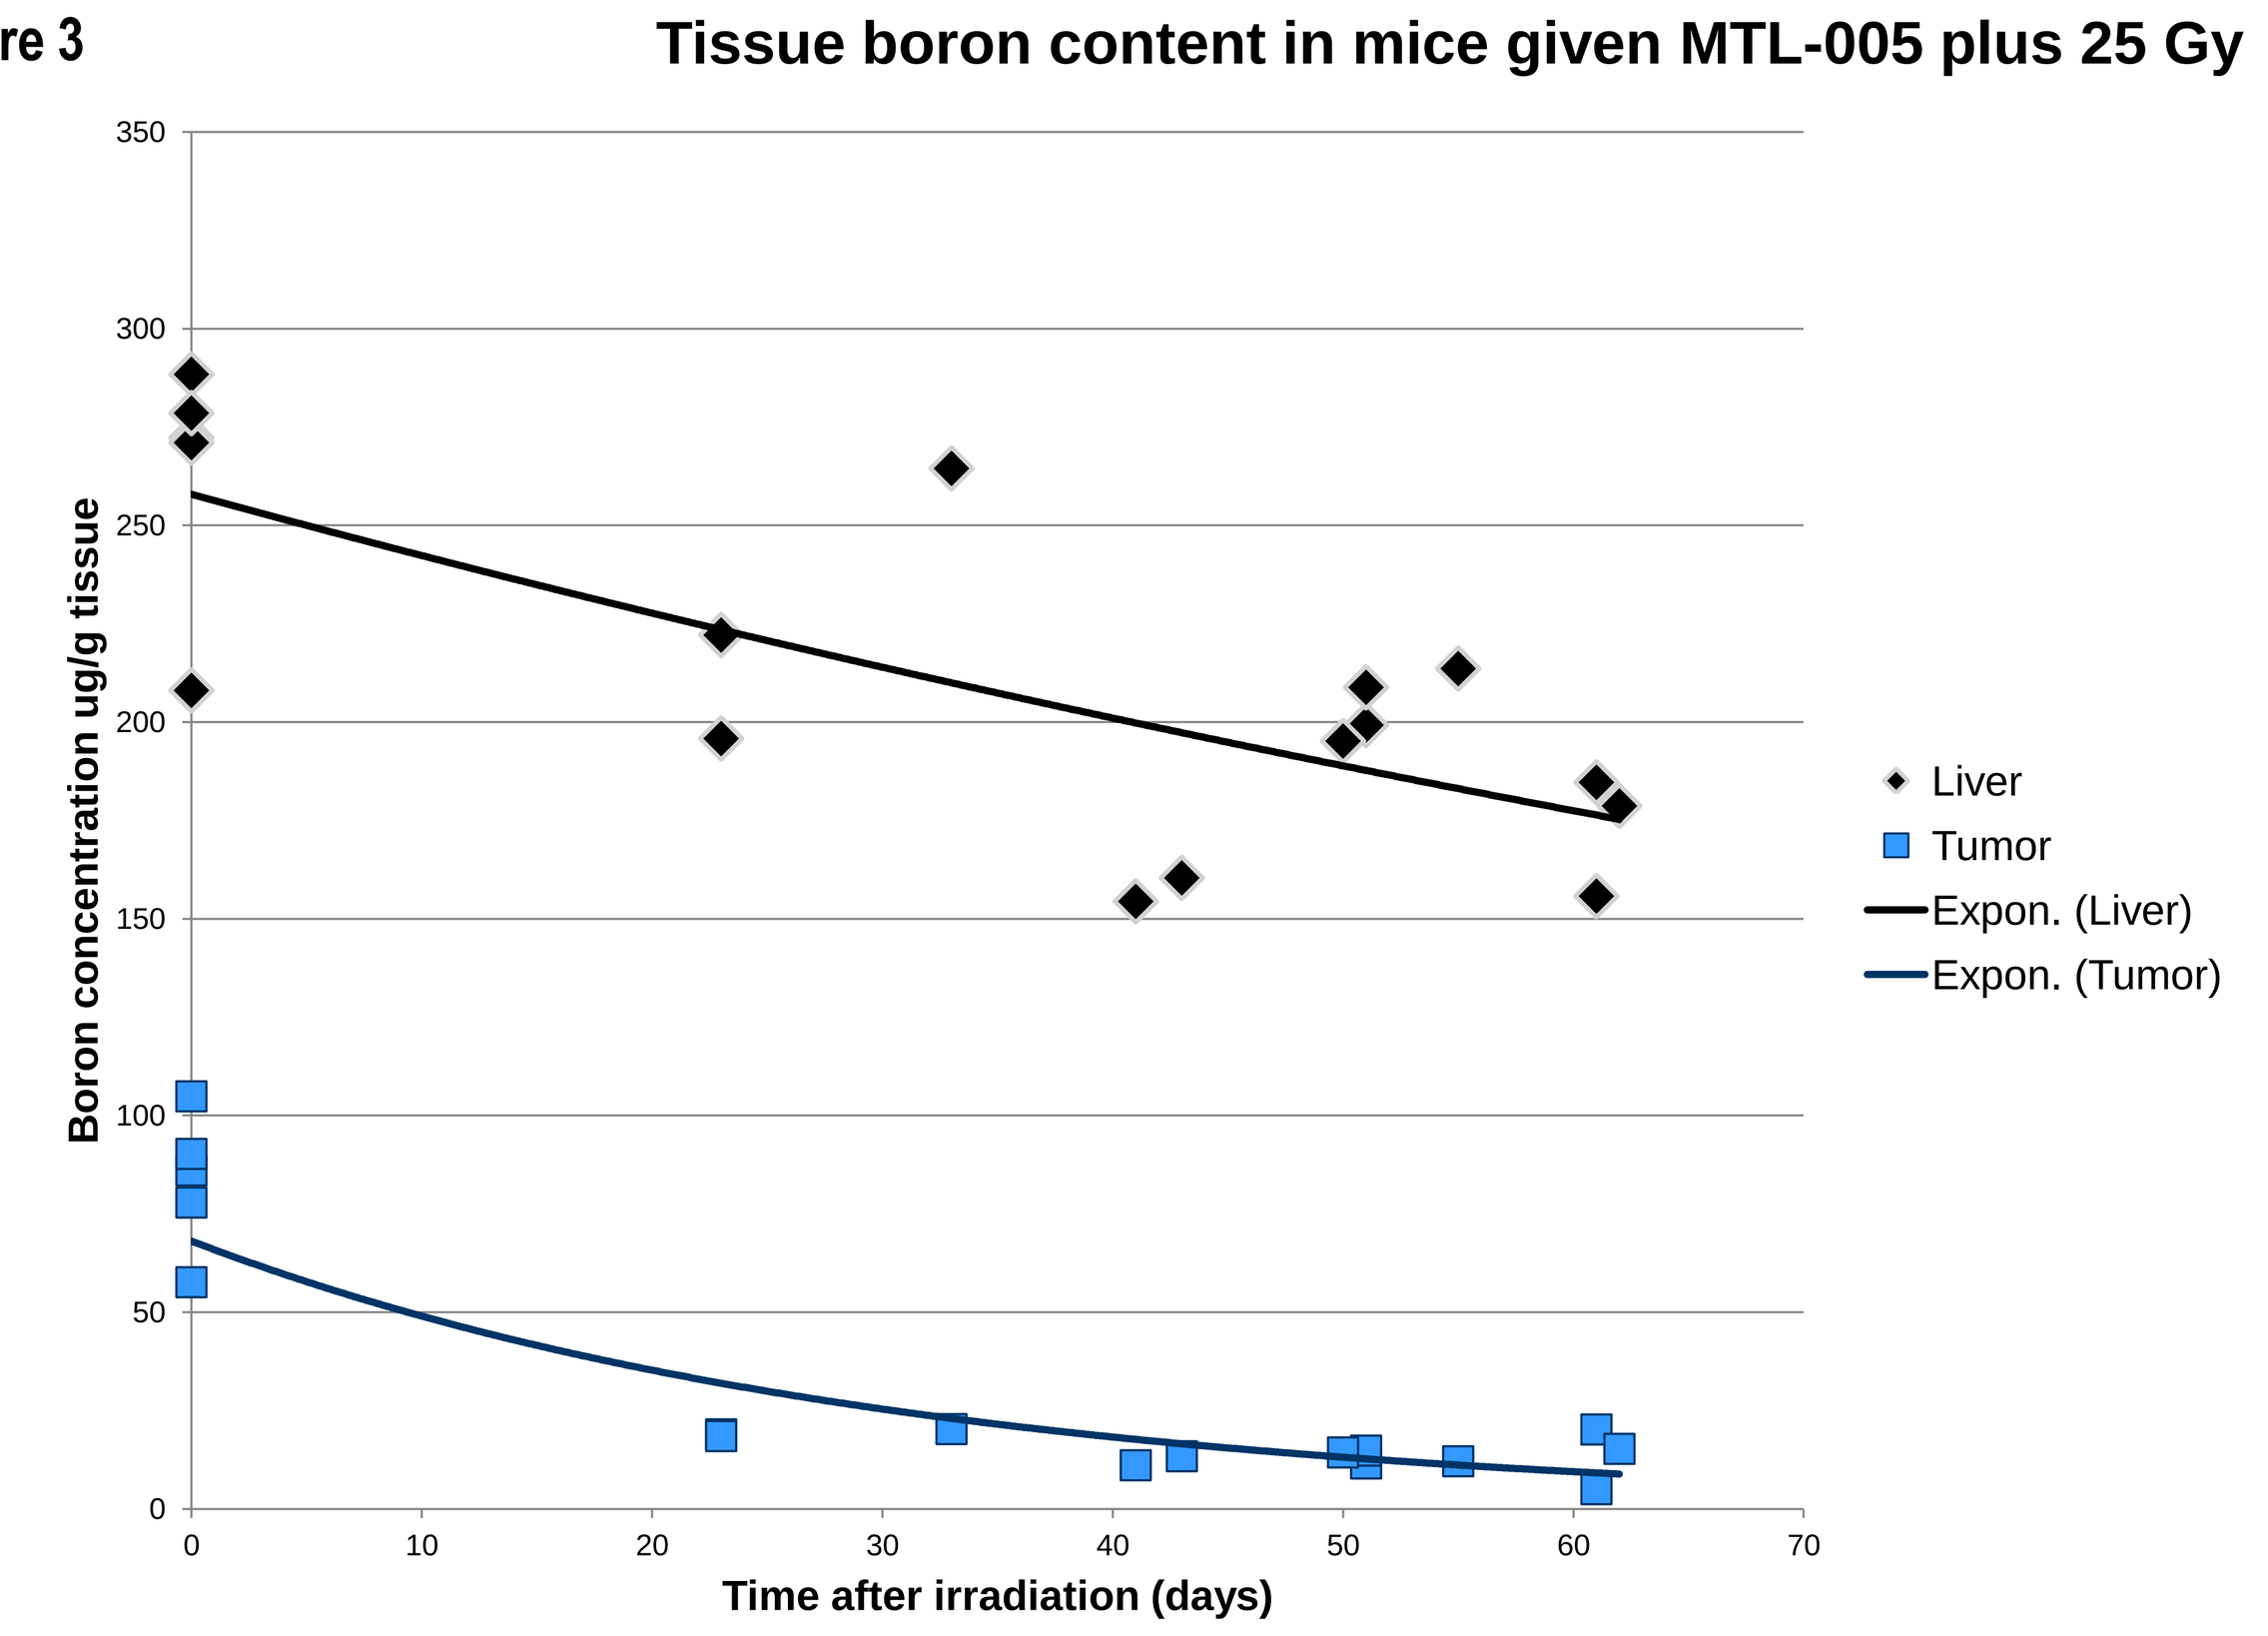
Tissue boron content in mice given MTL-005 plus 25 Gy
Boron concentration ug/g tissue
Time after irradiation (days)
Liver
Tumor
Expon. (Liver)
Expon. (Tumor)

Figure 4

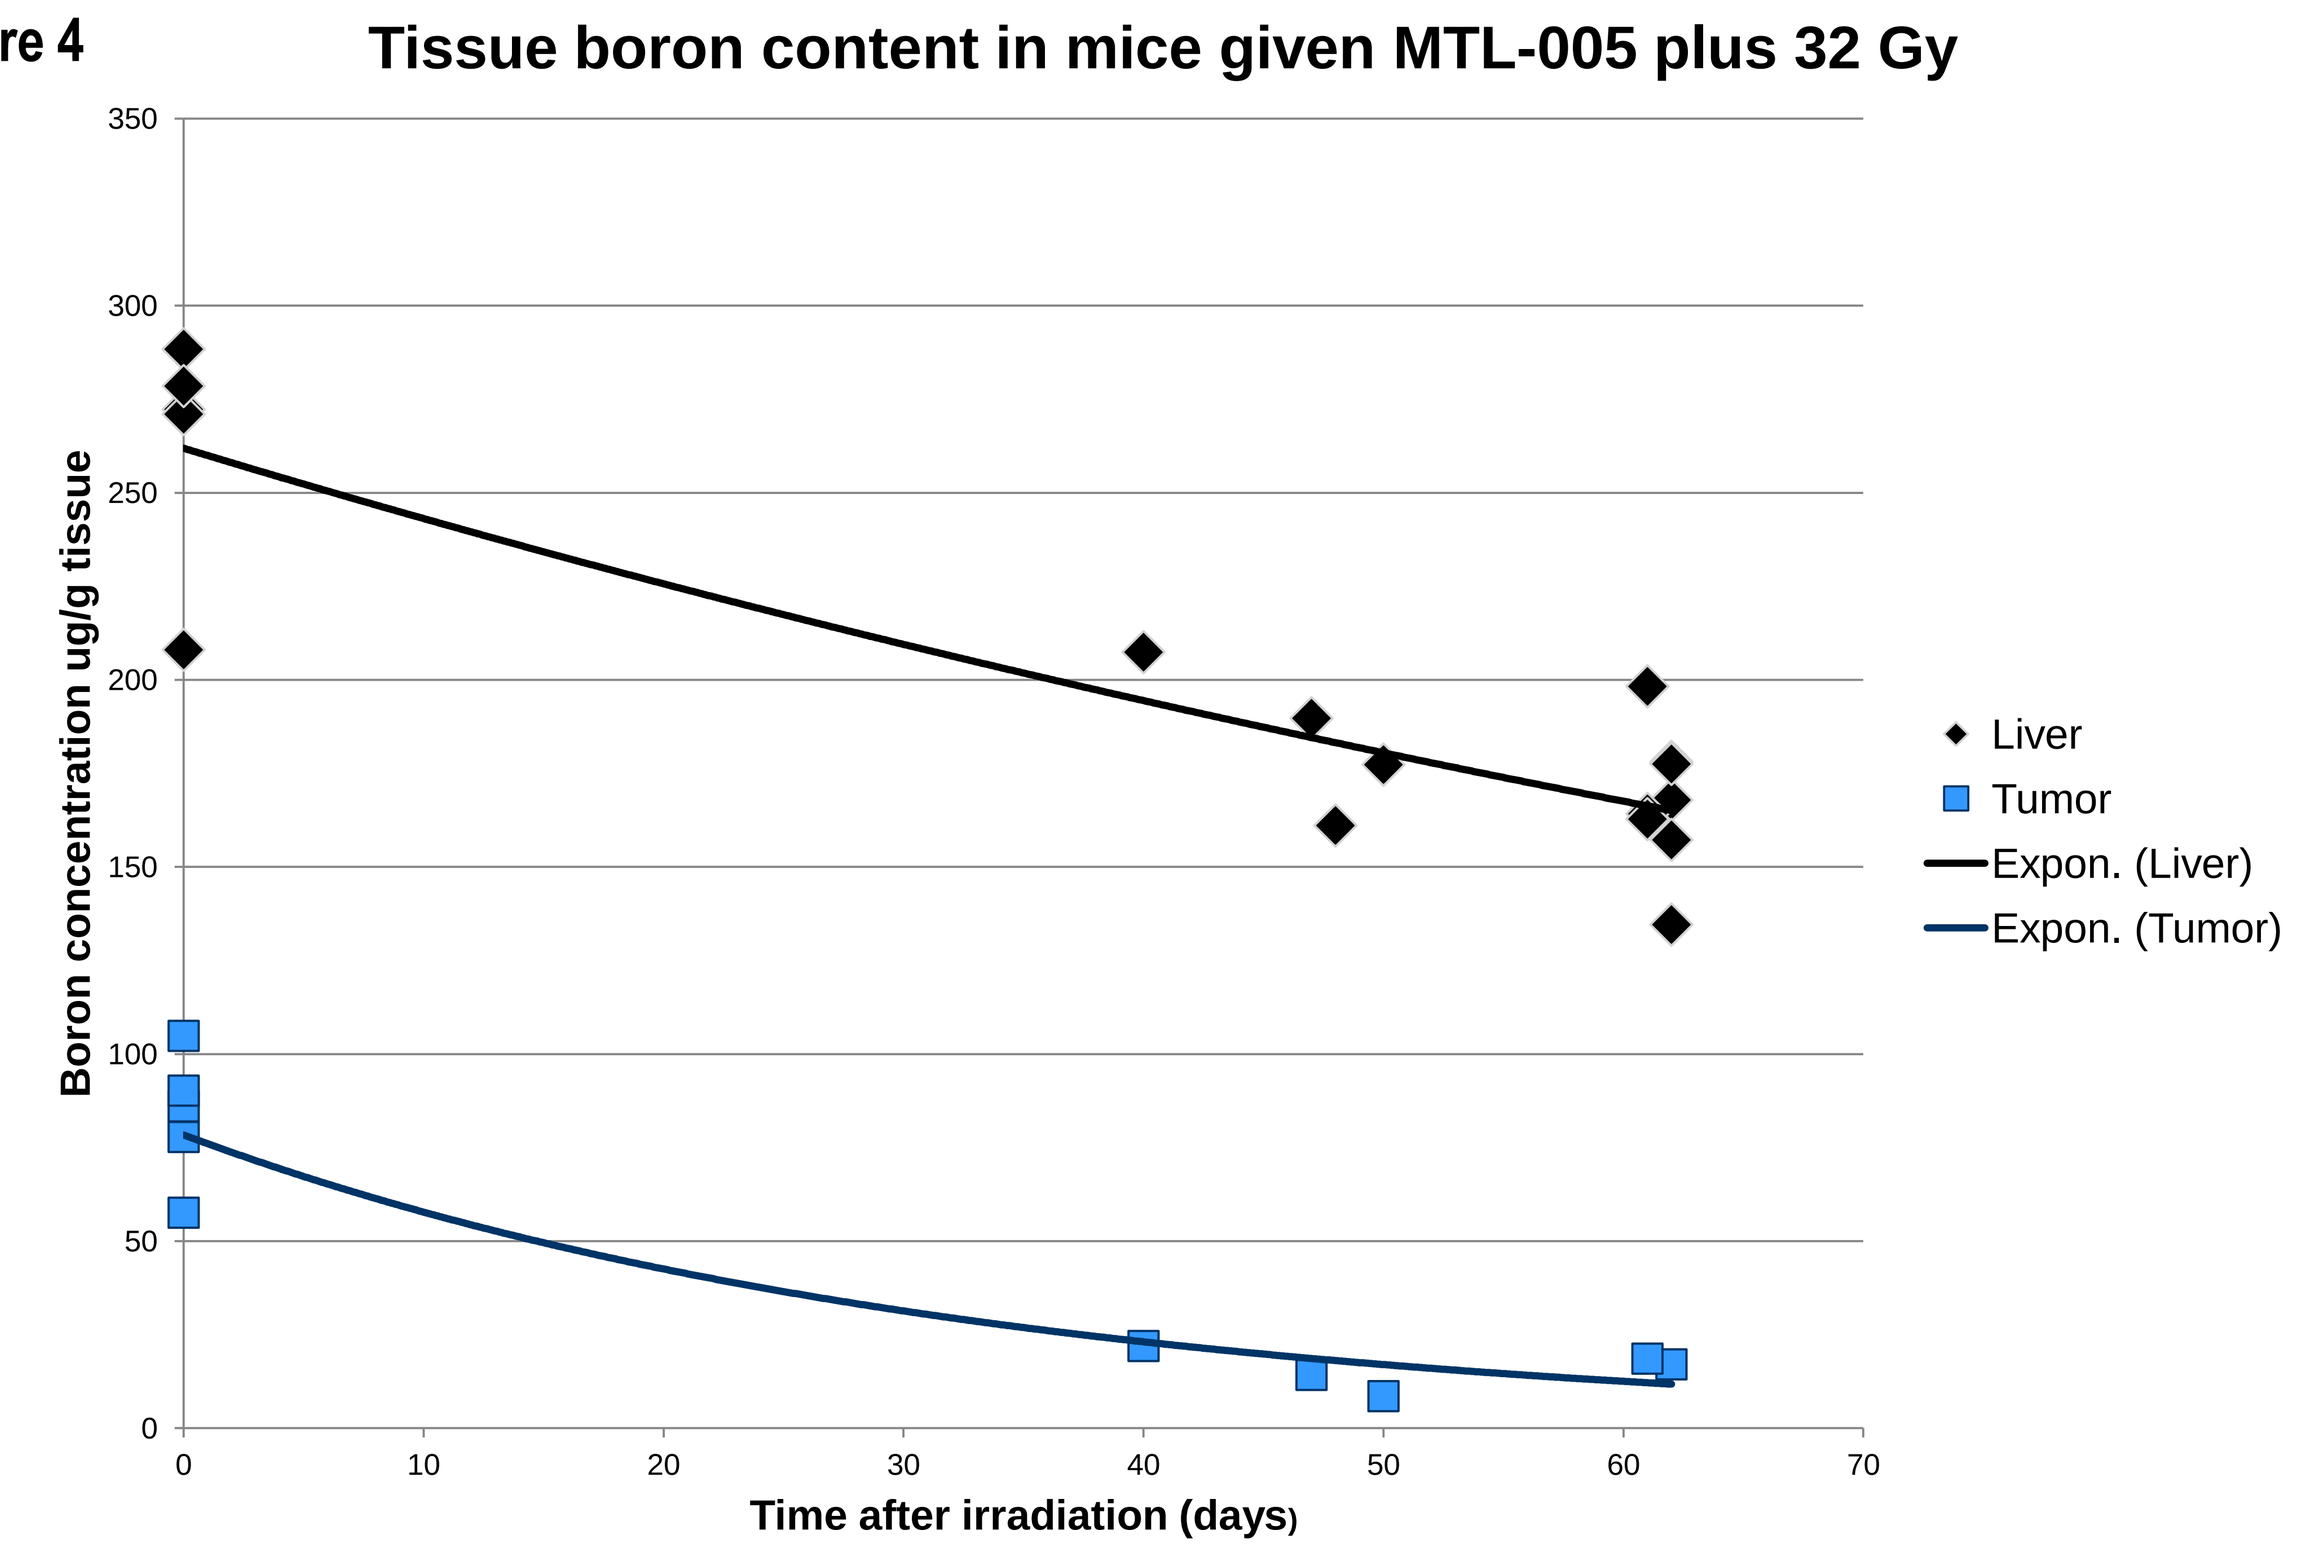
Tissue boron content in mice given MTL-005 plus 32 Gy
Boron concentration ug/g tissue
Time after irradiation (days)
Liver
Tumor
Expon. (Liver)
Expon. (Tumor)

Conclusions

MTL-005 significantly enhanced the effectiveness of radiotherapy of a squamous cell carcinoma (SCCVII) implanted in mice, slowing tumour growth and increasing the number of controlled tumors. The number of controlled tumors was doubled in the presence of MTL-005 compared to radiation alone following a single dose of 32Gy. In contrast there did not appear to be a significant tumor control effect at 25 Gy, although tumor growth was delayed. MTL-005 remained at detectable levels in tumor up to 63 days post administration which was 62 days post irradiation. MTL-005 is worthy of further investigation. A Phase I clinical trial was initiated in the United Kingdom in Q4 2013.

References

1. KK Fu *et. al*. The influence of time sequence of cisplatin administration and continuous low dose rate irradiation on their combined effects on a murine squamous cell carcinoma. *Int. J Radiat Oncol Biol Phys* **1985**, 11: 2119-2124
2. Y Saito *et al*. Effect of radiation and paclitaxel on p53 expression in murine tumors sensitive or resistant to apoptosis induction. *Int J Radiat Oncol Biol Phys* **1997**, 38: 623-631
3. Barth RF *et al* Determination of boron in tissues and cells using direct current plasma atomic emission spectroscopy. *Anal Chem* **1991**, 63: 890- 893
4. Fairchild RG et al Microanalytical techniques for boron analysis using the $^{10}B(n,\alpha)^7Li$ reaction *Med Phys* **1986**, 13, 50-56
5. Klevenhagen SC *et al* The IPEMB code of practice for the determination of absorbed dose for x-rays below 300 kV generating potential (0.035 nm Al-4 mm Cu HVL; 10-300kV generating potential) *Phys Med Biol* **1996** 41: 2605-2625

TEMPLATE DESIGN © 2008
www.PosterPresentations.com